THE USE OF A RESPONDER ANALYSIS TO IDENTIFY CLINICAL MEANINGFUL DIFFERENCES IN PATIENTS SUFFERING FROM CHRONIC URTICARIA FOLLOWING A 4-WEEKS TREATMENT WITH RUPATADINE 10- AND 20 mg

A. Gimenez-Arnau [1], I Pérez [2], E Donado [2], RM Pujol-Vallverdú [1], Arnaiz E [2], I Izquierdo [2]
[1]Servicio de Dermatología. Hospital del Mar, IMAS. Universitat Autònoma de Barcelona.
[2]Departamento de Desarrollo Clínico y Asesoria Médica , Uriach Pharma, Barcelona

BACKGROUND

Chronic urticaria (CU) is defined by spontaneous wheals long-lasting more than 6 weeks.[1] Conventionally, CU was categorized as idiopathic (CIU) if not apparent etiology was considered. It is a common skin condition with a 0.5% worldwide lifelong prevalence across different populations, affecting between 0.1% and 3% people in Europe and in USA. CU get worse the Quality of Life (QoL), primarily as a result of sleep disruption, energy loss, fatigue, social isolation and emotional/sexual disturbances.[2] This condition follows a chronic course with spontaneous remission and relapses for several years.
The symptoms of chronic urticaria, included oedema, erythema and pruritus, are primarily associated with histamine release from dermal mast cells. Oral H_1-receptor inverse agonists (H_1 antihistamines) are the treatment of choice.[3]
According with the EAACI/GA²LEN/EDF guidelines for urticaria management and diagnosis, the new no-sedating antihistamines are the first line of treatment for chronic urticaria.[4]
Other mast cell mediators including eicoanoids, cytokines, proteases, cytokines and platelet activating factor (PAF) are also involved in wheal development. PAF and histamine have mutually complementary activities *in vivo* [5]. Dual blockade of these mediators is likely to be more effective treatment strategy for chronic urticaria.

Rupatadine is a selective long-lasting histamine H1-receptor inverse agonist, currently authorized in European Community for the treatment of chronic idiopathic urticaria [6,7] and allergic rhinitis [8] (See fig 1). This drug has shown both antihistamine and anti-PAF effects through its interaction with specific receptors.[9] Only few others H1 antihistamines showed some marginal anti-PAF effect.[10]

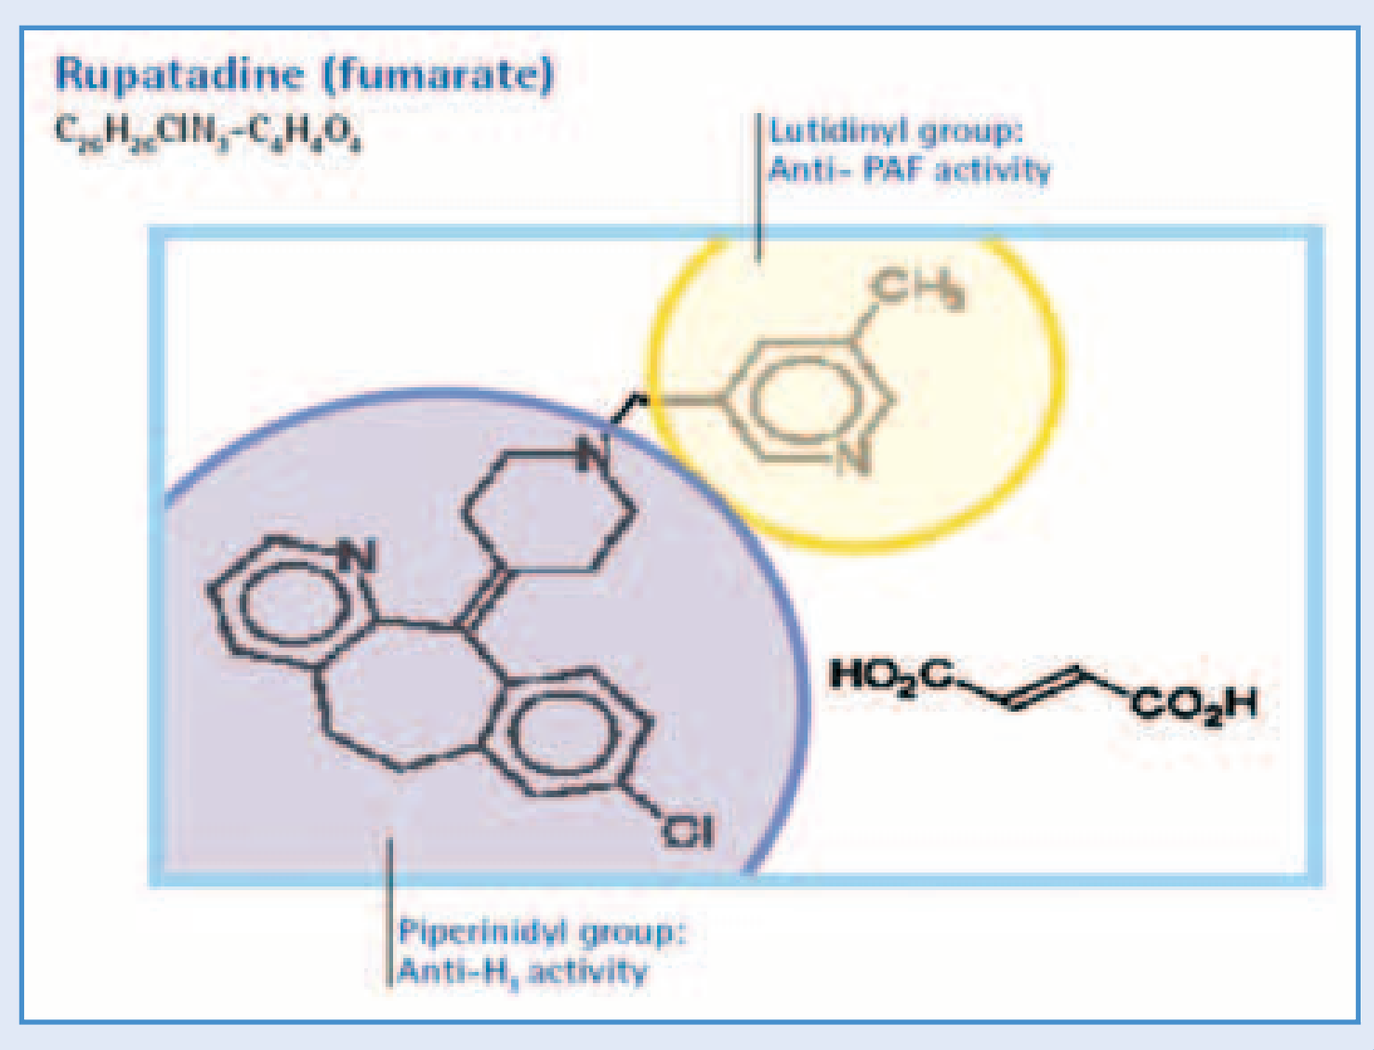
Rupatadine (fumarate)
$C_{26}H_{26}ClN_3 \cdot C_4H_4O_4$
Lutidinyl group:
Anti- PAF activity
Piperinidyl group:
Anti-H_1 activity

Fig 1. Chemical structure

PURPOSE OF THE STUDY

A responder analysis in chronic idiopathic urticaria (CIU) patients was performed by means of a pooled analysis, in order to strengthen the clinical results obtained in previous trials with rupatadine 10 and 20 mg.

MATERIAL AND METHODS

The pooled data from two randomised, double blind, placebo-controlled, 4-week multicentre studies were used for this analyses. The first trial was a dose-ranging study comparing the efficacy and safety of rupatadine 5-, 10- and 20 mg once daily or placebo in 248 patients [6]. The second study compared the efficacy of rupatadine 10 and 20 mg, once daily with placebo, in 334 CIU patients [7].
In both trials, patients were included if they suffered moderate to severe CIU as active CIU (score ≥ 2 labeled as moderate pruritus) for at least 3 days during the week before inclusion, with a total score of active CIU ≥ 6 and documented history of active CIU (urticaria wheals) with or without an associated angioedema for at least three days per week over the last 6 weeks prior the Screening Visit.
Response rates are defined in Table 1.
The statistical pooled responder's analyses employed two logistic models. Model 1 extracted effects for study, treatment and study-by-treatment interaction. In the absence of heterogeneity (not significant interaction between study and treatment), the final model extracted only effects for study and treatment. Heterogeneity was not found in any of the responder's analysis. The values of percentage of improvement are shown in tables and figures as raw values, whereas the p value is obtained from the final logistic model.

Table 1. Summary of responder criteria

Criteria	Definition	Based on
>50% Reduction from baseline	Percentage of patients who achieved an improvement in symptom score of at least 50% from baseline in each treatment group	% Change from baseline
> 75% Reduction from baseline	Percentage of patients who achieved an improvement in symptom score of at least 75% from baseline in each treatment group	% Change from baseline
Reduction greater than those observed	Percentage of patients over the Lower 95% Confidence Interval with placebo for the mean change from baseline in placebo group	Absolute Change from baseline

RESULTS

538 patients were treated with placebo, rupatadine 10 and 20 mg (ITT population). (Table 2)
Figures 2, 3 and 4 shows the percentage of patients that obtained a >50%, >75% and LCLM of scores improvement, Mean Pruritus Score (MPS), Mean Number of Wheals (MNW) and Means Total Symptoms Score (MTSS = MPS + MNW) after 28 days.; p<.002.

Table 2. Number of patients with CIU included in clinical trials of rupatadine

Study	Placebo	Rupatadine 10 mg	Rupatadine 20 mg	Total
Phase II ICO2RUP/II/02	69	73	67	209
Phase III ICO10RUP/3/04	111	110	108	329
Total	180	183	175	538

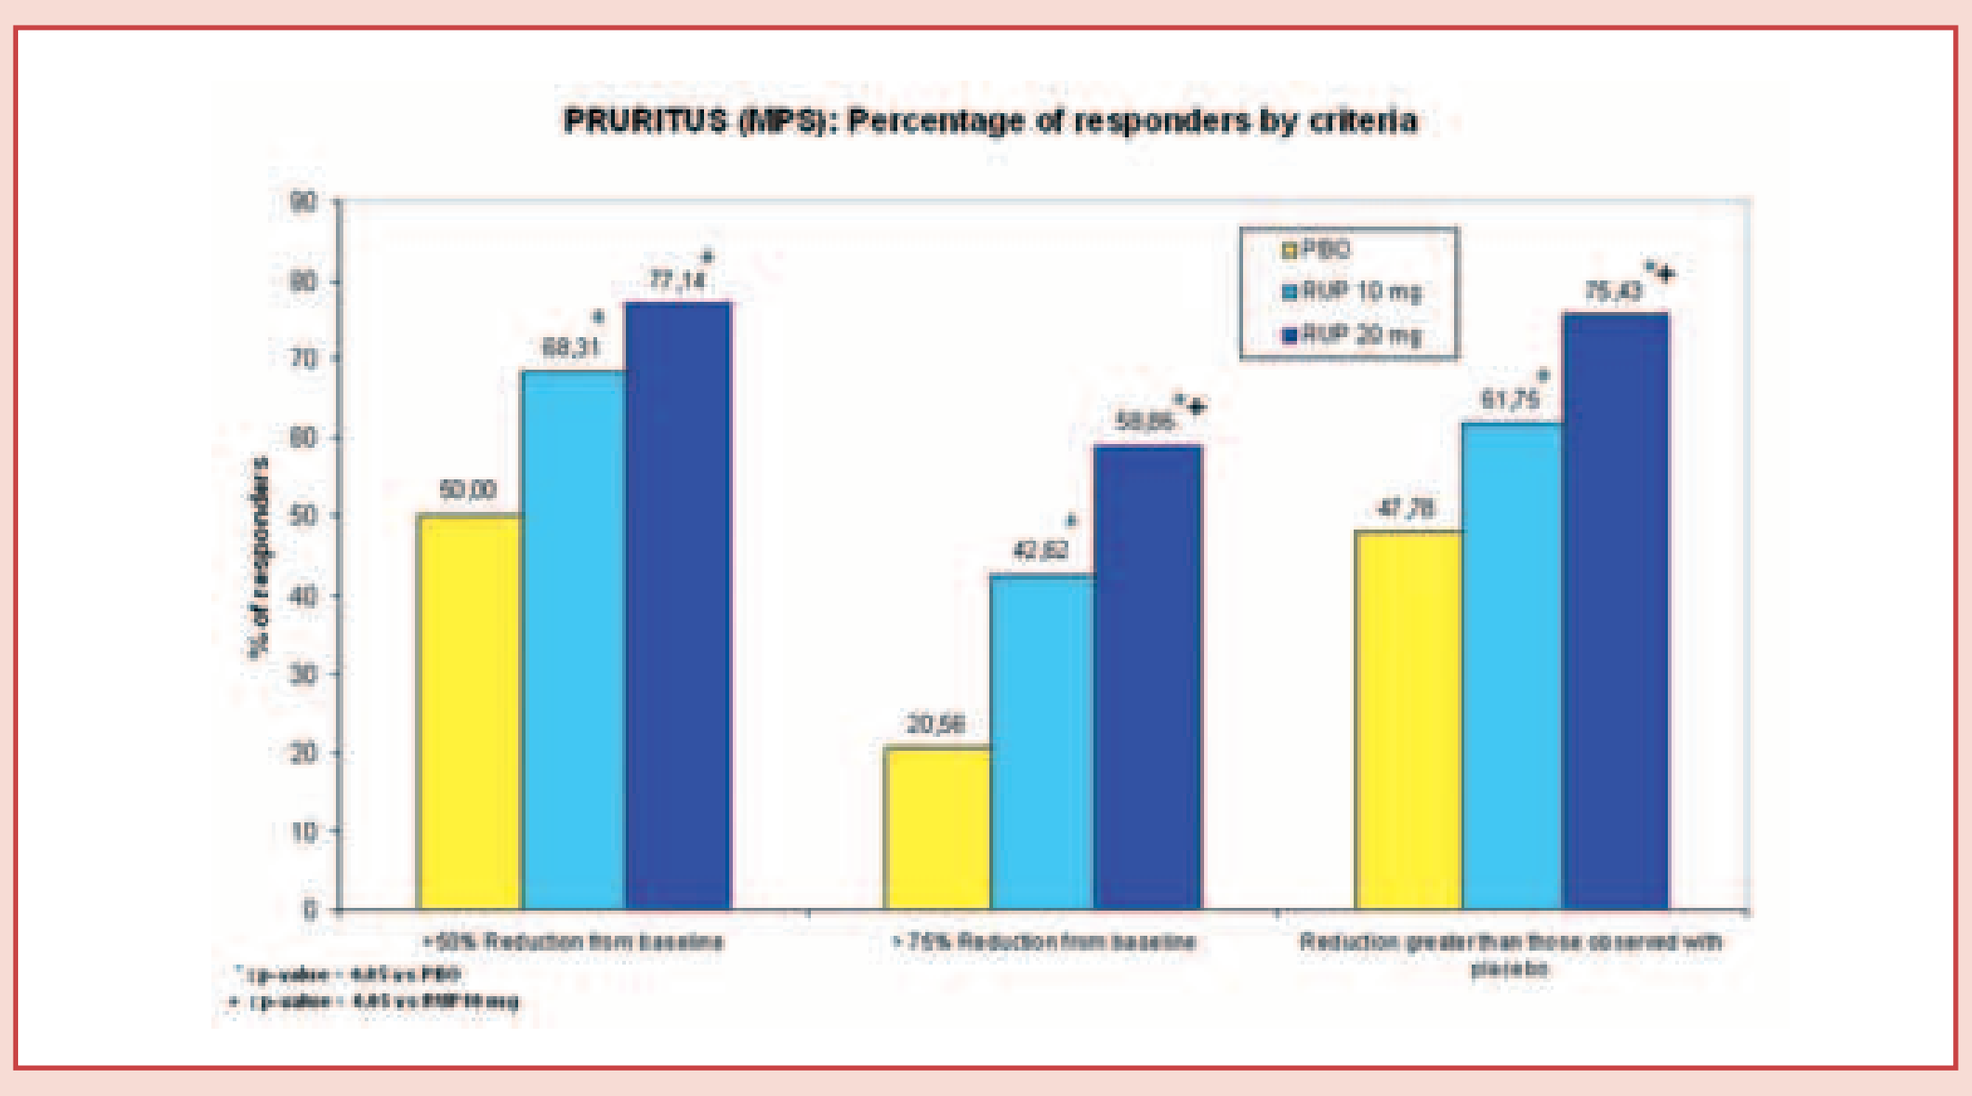
PRURITUS (MPS): Percentage of responders by criteria
% of responders
PBO
RUP 10 mg
RUP 20 mg
50,00
68,31
77,14
20,56
42,62
58,86
47,78
61,75
75,43
>50% Reduction from baseline
>75% Reduction from baseline
Reduction greater than those observed with placebo

Fig 2. MPS

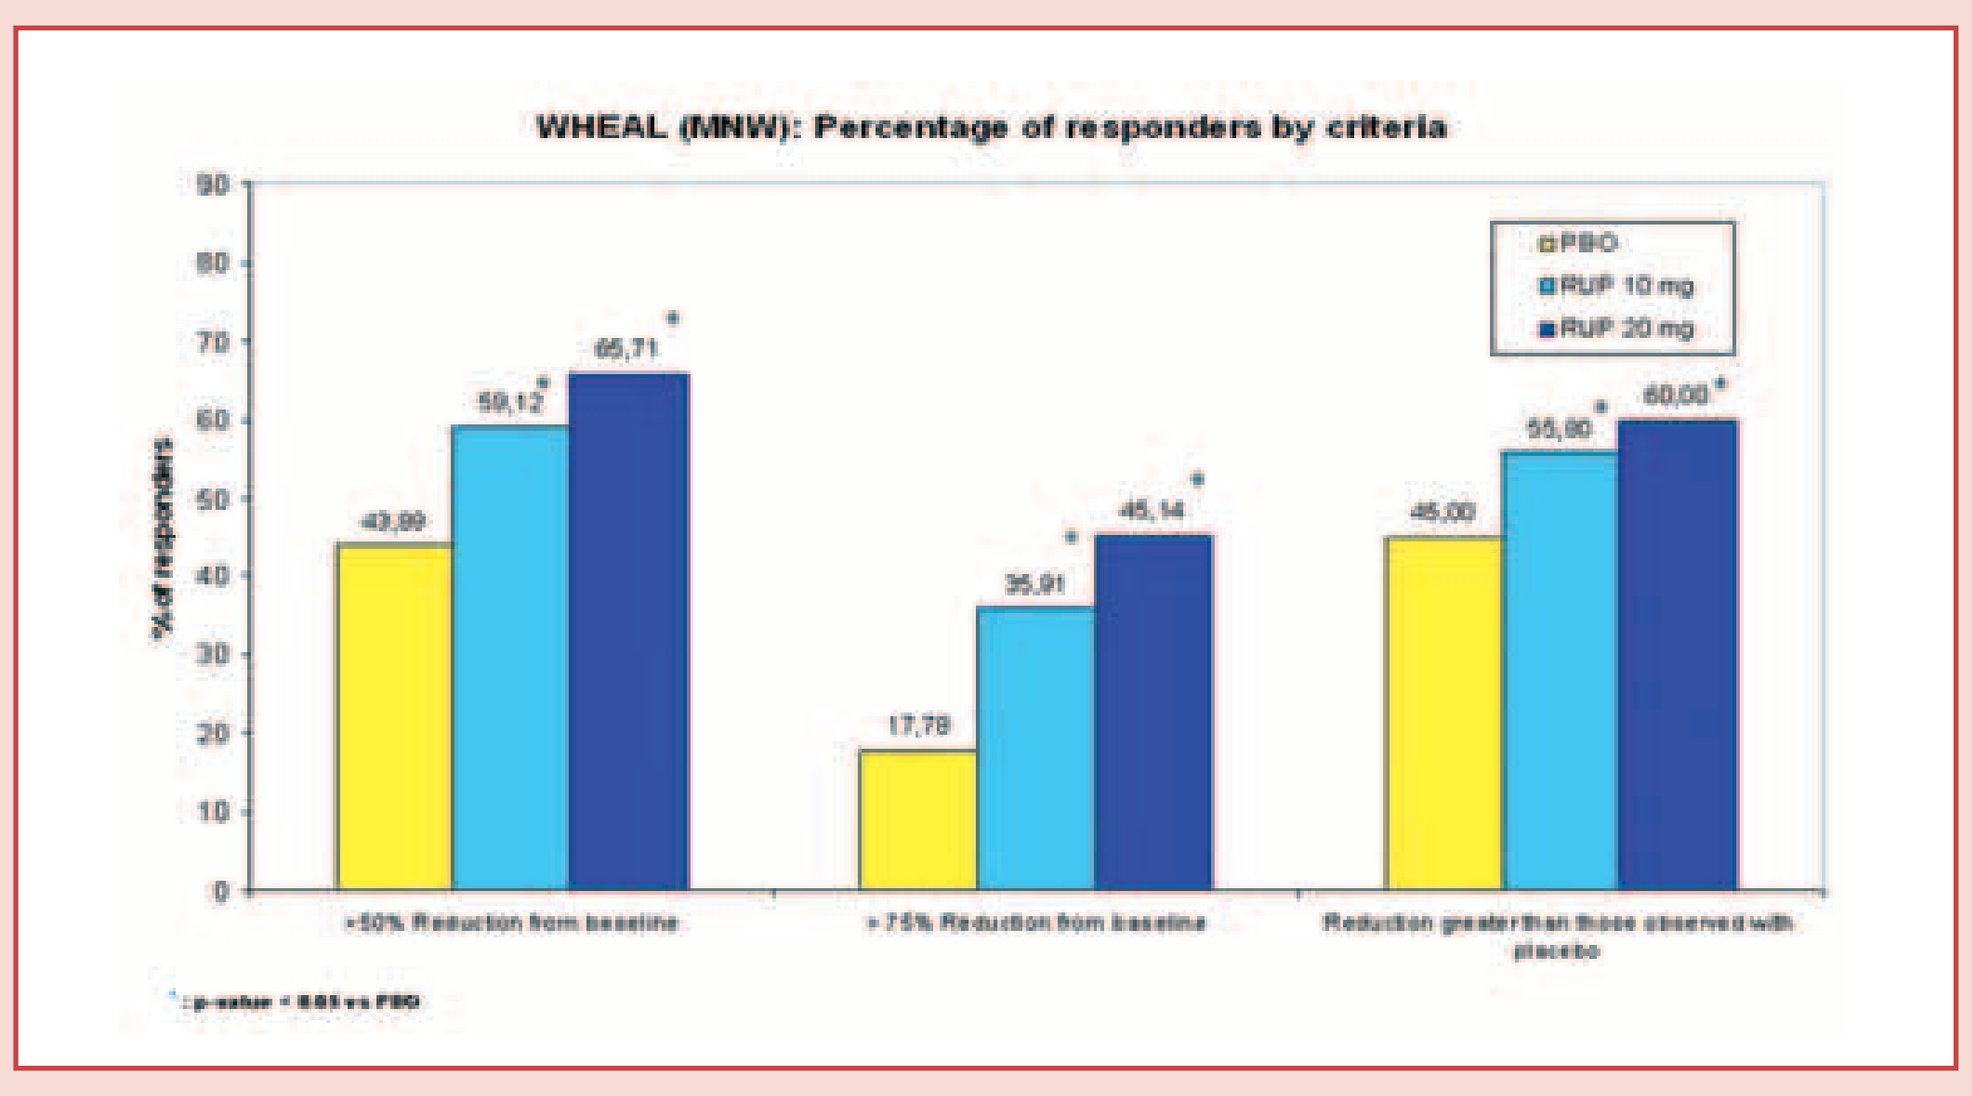
WHEAL (MNW): Percentage of responders by criteria
% of responders
PBO
RUP 10 mg
RUP 20 mg
43,89
59,12
65,71
17,78
35,91
45,14
45,00
55,80
60,00
>50% Reduction from baseline
>75% Reduction from baseline
Reduction greater than those observed with placebo

Fig 3. MNW

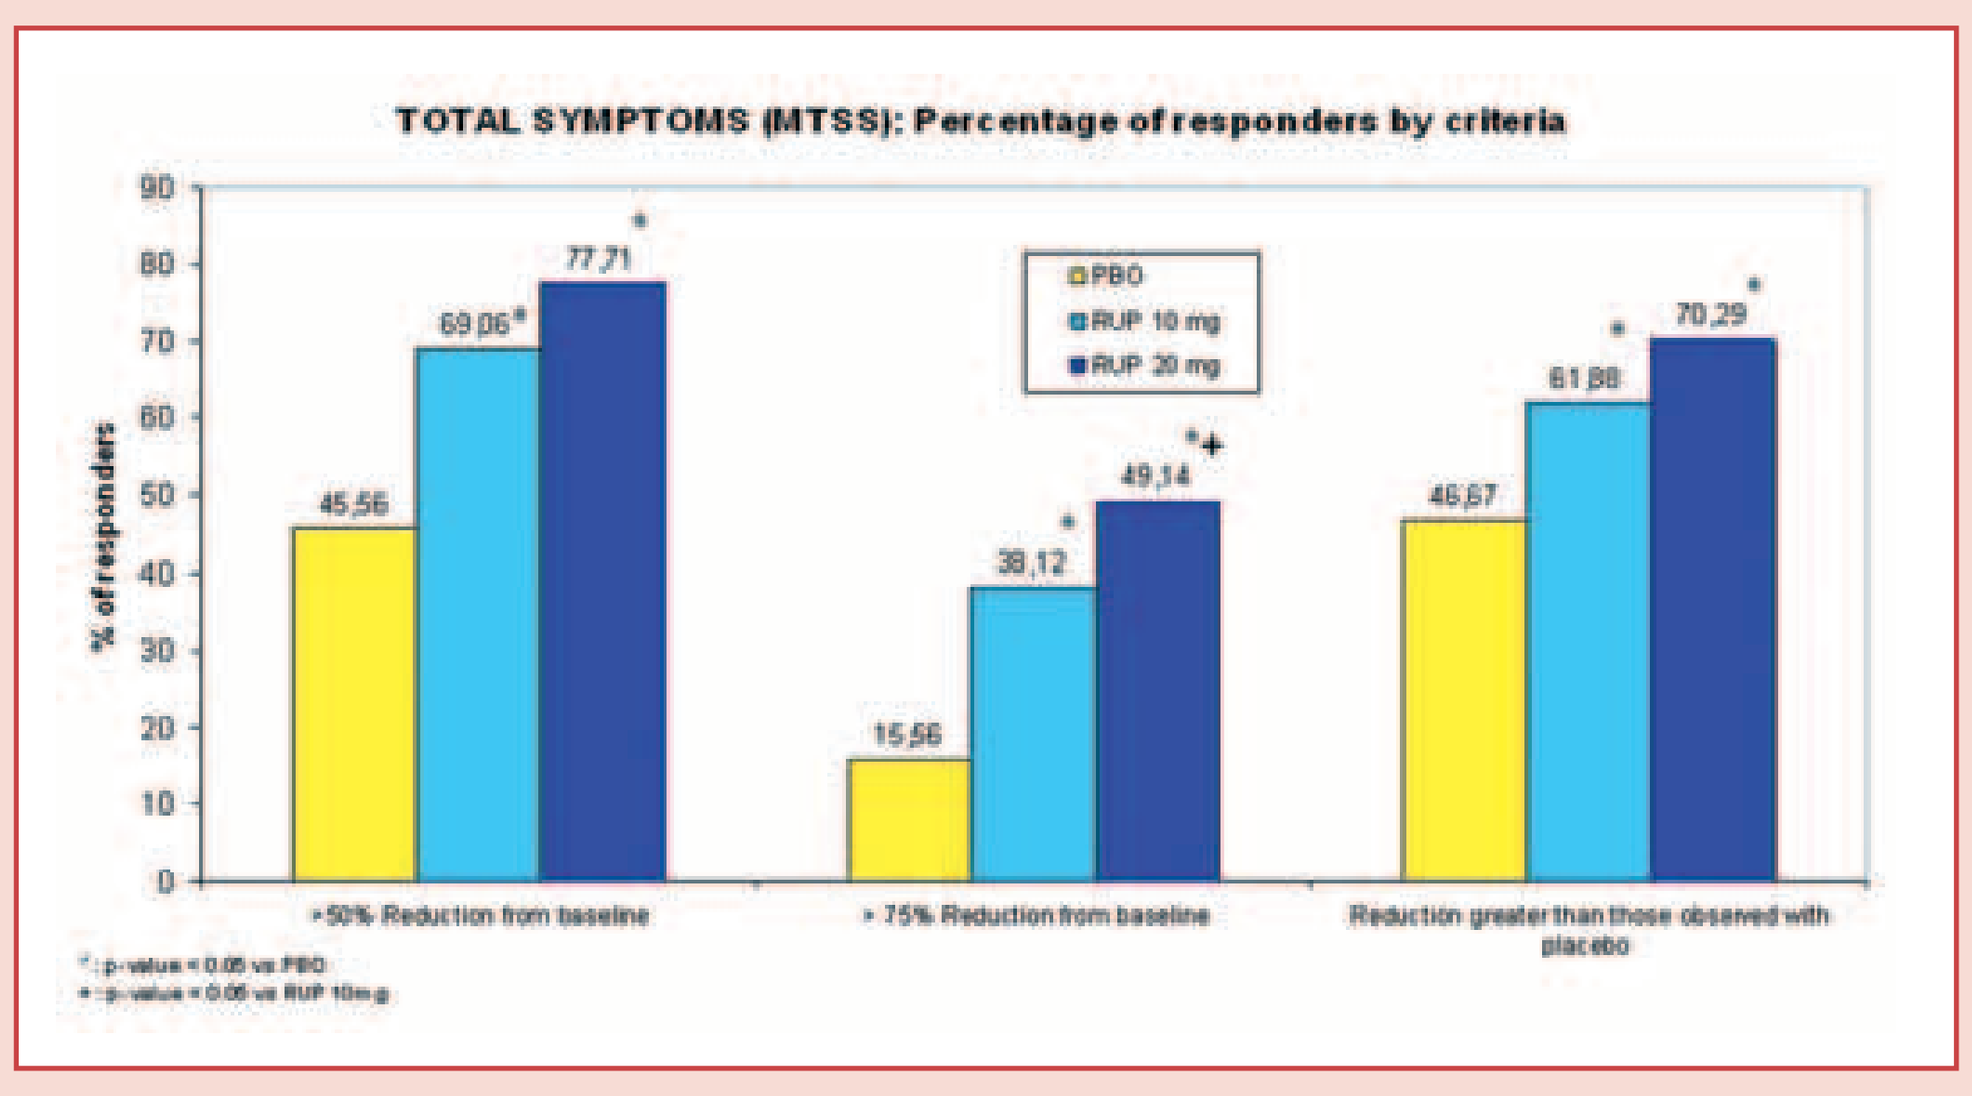
TOTAL SYMPTOMS (MTSS): Percentage of responders by criteria
% of responders
PBO
RUP 10 mg
RUP 20 mg
45,56
69,06
77,71
15,56
38,12
49,14
46,67
61,88
70,29
>50% Reduction from baseline
> 75% Reduction from baseline
Reduction greater than those observed with placebo

Fig 4. MTSS

DISCUSSION

CU treatment is difficult. Eliciting stimuli must be investigated and controlled. Mast cell directed therapy includes corticosteroids, cyclosporin A or phototherapy. Target organ or symptomatic therapy is normally done with H_1-receptor antagonists. Because of the good safety profile, second-generation antihistamines are considered the first line symptomatic treatment for chronic urticaria. But there is any consensus on which is the percentage of clinical improvement that we must expect using the recommended dose for each antihistamine in a defined period of time. Most experts agree with the fact that higher dosages should be used as routine [4]. Some others experts recommend to initiate the treatment with the combination of more than one antihistamine (sedative with non sedative).[11]. A 50% of symptoms reduction after 28 days of treatment employing low dose of just one antihistamine is considered an adequate improvement.
This responder analysis, at different responses levels, showed that the efficacy of rupatadine 10 mg is statistically significant compared to placebo in all analyzed variables, consistent across all pre-defined responses ranges and clinically relevant as suggested by current dermatologist criteria. Using LCLM definition (a more restrictive criteria) similar results were observed. Rupatadine at 20 mg showed a higher improvements in all scores with significantly differences in 75% reduction of MPS and MTSS in comparison with rupatadine 10 mg.

CONCLUSION

1. The responder analysis ads strengthen to the rupatadine clinical trials results.
2. It would help the clinicians to evaluate the rupatadine response in CIU patients.
3. This study support the need to use higher doses of no sedating antihistamines in chronic urticaria.
4. A complete responder analysis should be considered in order to evaluate antihistamine therapeutic benefits.

ACKNOWLEGEMENTS

The authors would like to thank J. Uriach y Compañía S.A. (Barcelona, Spain) for its financial support to this study. This study was partially supported by the National Scientific Research Program of the Spanish Minister of Science and Technology.

REFERENCES

1. Greaves MW. Chronic Urticaria. N Engl J Med 1995; 322: 1767-1772.
2. O, Donnell BF, Lawlor F, Simpson J, Morgan M, Greaves MW. The impact of chronic urticaria on the quality of life. Br J Dermatol 1997; 136: 197-201.
3. Kozel MMA, Sabroe RA. Chronic Urticaria. Etiology, management and current future treatment options. Drugs 2004; 64: 2515-36.
4. Zuberbier T, Bindslev-Jensen C, Canonica W, Grattan EH, Greaves MW, Henz BM, Kapp A, Kozel MMA, Maurer M, Merk HF, Schäfer T, Simon D, Vena GA, Wedi B. EAACI/ GA²LEN/ EDF guideline: management of urticaria. Allergy 2006; 61: 321-33.
5. Petersen IJ, Churck MK, Skov PS. Platelet-activating factor induces histamine release from human mast cells in vivo, which is reduced by local nerve blockage. J Allergy Clin Inmunol 1997; 99: 640-7.
6. Dubertret L, Zalupca L, Cristodoulo T, Benea V, Medina M, Fantin S, Lahfa M, Pérez P, Izquierdo I, Arnaiz E. Once-daily rupatadine improves the symptoms of chronic idiopathic urticaria: a randomised, double-blind, placebo-controlled study. Eur J Dermatol 2007; 17: 223-228.
7. Gimenez-Arnau A, Pujol RM, Ianosi S, Kaszuba A, Malbran A, Poop G, Donado E, Perez I, Izquierdo I, Arnaiz E. Rupatadine in the treatment of chronic idiopathic urticaria: a double-blind, randomized, placebo-controlled multicenter study. Allergy 2007; 62: 539-546.
8. Guadaño EM, Serra-Batlles, Meseguer J, Castillo JA, Molina M, Valero A, Picado C. Rupatadine 10 mg and Ebastine 10 mg in seasonal allergic rhinitis: a comparison study. Allergy 2004; 59: 766-771.
9. Merlos M, Giral M; Balsa D, Ferrando Q, Queralt M, Puigdement A, García-Rafanell J and Forn J. Ruptadine, a new potent, orally active dual antagonist of histamine and platelet-activating factor (PAF). J Pharmacol Exp Ther 1997; 280: 114-121.
10. Juhlin L, Rihoux JP. Effect of ceterizine on cutaneous reactions to PAF, kallikrein and serum in patients with chronic urticária. Acta Derm Venereol 1990; 70: 151-152.
11. Sabroe RA, Greaves MW. Chronic idiopathic urticaria with functional autoantobodies: 12 years on .Br J Dermatol 2006; 154: 813-819.

hospitaldelmar

C S B Consorci Sanitari de Barcelona
IMAS
Institut Municipal d'Assistència Sanitària