WHEAL AND SKIN GENE EXPRESSION IN CHRONIC SPONTANEOUS URTICARIA

Giménez-Arnau A[1], Curto-Barredo L[1], Nonell L[2], Puigdecanet E[2], Alloza L[2], Santamaria-Babi L[3], Pujol R M[1]

[1]Department of Dermatology Hospital del Mar, Universitat Autònoma de Barcelona. [2]Microarray Analysis Service, Institut Mar d'Investigacions Mèdiques. [3]Translational Immunology. Department of Physiology and Immunology, Universitat de Barcelona

INTRODUCTION AND OBJECTIVE

Phenotypic and genotypic profiling of Chronic Spontaneous Urticaria (CSU) an IgE-associated disease still is an unmet need. Some of the mechanisms involved in mast cell activation and the role of the released mast cell mediators in CSU are well known. However the knowledge about the genetic susceptibility and genes expressed in the wheal and the non lesional skin of CSU patients remain limited. The primary purpose of this study was to identify and characterize the gene expression profile in lesional (wheal) and non lesional skin in a series of patients suffering from severe CSU.

METHODOLOGY

Twenty patients suffering CSU were studied (14 -70% women and 6-30% man) with a mean age of 51.55 ± 15.7 (SD) years [20-81]. The mean duration of the disease was 4.9 ± 9 (SD) years [3m-40y] and the mean UAS 7 was 33.6 [0-42]. The control group showed a ratio female: male of 1:1 (5 women, 5 man) and the mean age was 27.5 ± 5.4 (SD) years. Cutaneous samples were obtained from a new wheal long-lasting 1 to 3 hours and from non-lesional skin. Gene analysis using Agilent whole human genome oligomicroarrays 8x60v2 (one-color) and a bioinformatics study based in the raw data was performed. Ingenuity Pathway Analysis (IPA) was used as first step for functional analysis. All results lists were loaded and a core analysis was done. The following comparisons were addressed: non lesional patient skin vs healthy skin from healthy donor and wheal vs non lesional skin. Given that there were statistically significant differences in age between patients and healthy controls, the variable age was added to the linear model in order to correct differences between those groups. This was performed with *limma* package (Smyth 2004). Based on technical recommendations (adjusted P value of <0.05 and a Log fold change of 1,2) and an accurate assessment of the genes up or down regulated according mast cell biology, wheal pathogenesis or urticaria knowledge, a number of genes were analyzed by quantitative real time PCR (Q-PCR) using Taqman Low Density Array (TLDA) platform. Up and down regulated genes were included analyses. A set of independent sample also was studied.

RESULTS

Patient non lesional skin vs Healthy skin from healthy donor

376 transcripts were found to be differentially expressed. An extended list of 799 transcripts was obtained only filtering by an adj.p.value <0.05. 39 genes were selected based on the adj p values <0.05 and logFC of 1 for validation using qPCR, form those, 31 were confirmed (80%).

Table 1. Some relevant genes confirmed by q-PCR in the comparison of non lesional skin vs healthy skin

Complete name of the gen	logFC	P adj Value
Small proline-rich protein 2E	4,41	0,00009
Late cornified envelope 2D	4,25	0,00002
Corneodesmosin	4,05	0,00001
Matrilin 4	3,34	0,00002
Filaggrin family member 2	3,24	0,00002
Desmocollin 1	2,77	0,00011
Arginase 1	2,61	0,00007
CD1a molecule	2,5	0,00001
Interleukin 37	2,41	0,00075
Fc frag of IgG binding protein	2,27	0
Interleukin 36, gamma	1,81	0,00015
SPINK5	1,71	0,00001
Keratin 10	1,63	0,00014
Epiplakin 1	1,6	0,00072
Retinol dehydrogenase 12 (all-trans/9-cis/11-cis)	1,59	0,00026
Filaggrin	1,43	0,00156
Repetin	1,26	0,00002
Cytokine-inducible SH2-containing protein	-2,63	0

Figure 1. Biological functions corresponding to the genes over or under expressed in non lesional skin vs healthy skin. Genes that were confirmed by Q-PCR

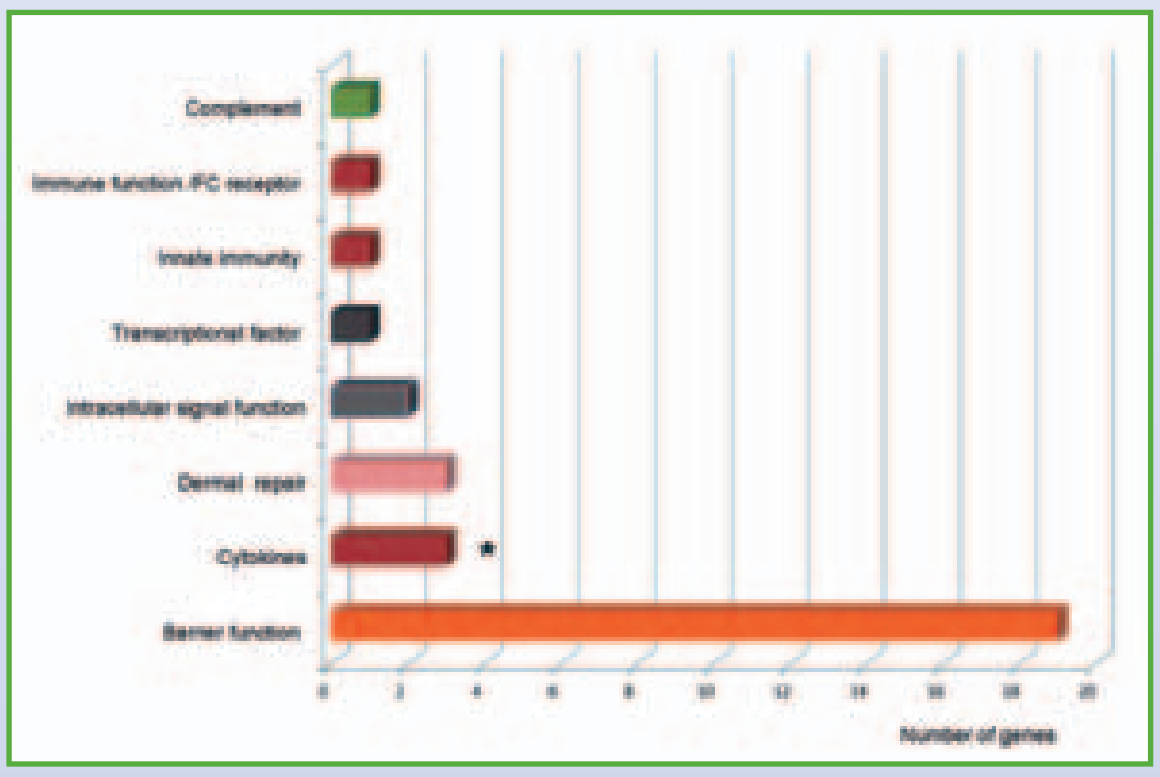
Complement
Immune function /FC receptor
Innate immunity
Transcriptional factor
Intracellular signal function
Dermal repair
Cytokines
*
Barrier function
Number of genes

* CISH is less expressed in no lesional skin

Wheal vs Non lesional skin from patients

To detect differences between wheal and healthy skin from patients, two analyses were conducted. In a first approach, an unpaired analysis of all available samples was performed, finding 713 differentially expressed transcripts (8,140 the extended list before filtering by logFC). The second analysis was performed on the 9 paired samples, obtaining a total of 700 differentially expressed ids (7,952 the extended list before filtering by logFC). From these two lists, 549 were coincident (78%). **142 genes were checked for confirmation using qPCR, from those, 103 were confirmed (73%).**

Table 2. Relevant genes confirmed by q-PCR in the comparison of wheal skin vs non lesional skin

Complete name of the gen	logFC	P adj Value
FOSB	3,93	0,00005
S100 calcium binding protein A9	3,38	0,00003
ADAMTS4	3,26	0,00003
Interleukin 6 (interferon, beta 2)	3,23	0,00005
Interleukin 1, beta	2,76	0,00005
Chemokine (C-C motif) ligand 2	2,47	0,00001
Formyl peptide receptor 1. G protein	2,46	0,00014
Selectin	2,43	0,00007
SERPINB3	2,38	0,00092
Activating transcription factor 3 (2 times)	2,29	0,00044
CYR61	2,24	0,00015
Peptidase inhibitor 3, skin-derived	2,09	0,00018
Involucrin	2,08	0,00008
Hyaluronan synthase 3	2,03	0,00017
Keratin 6C	1,93	0,00032
Prostaglandin-endoperoxide synthase 2	1,58	0,00003
Superoxide dismutase 2, mitochondrial	1,35	0,00143
ICAM1	1,29	0,00039
Aquaporin 3 (Gill blood group)	1,22	0,00007
Plasminogen activator, urokinase receptor	1,14	0,0013
Platelet-activating factor receptor	1,06	0,0017

Figure 2. Biological functions corresponding to the genes over or under expressed in wheal skin vs non lesional skin. Genes that were confirmed by Q-PCR

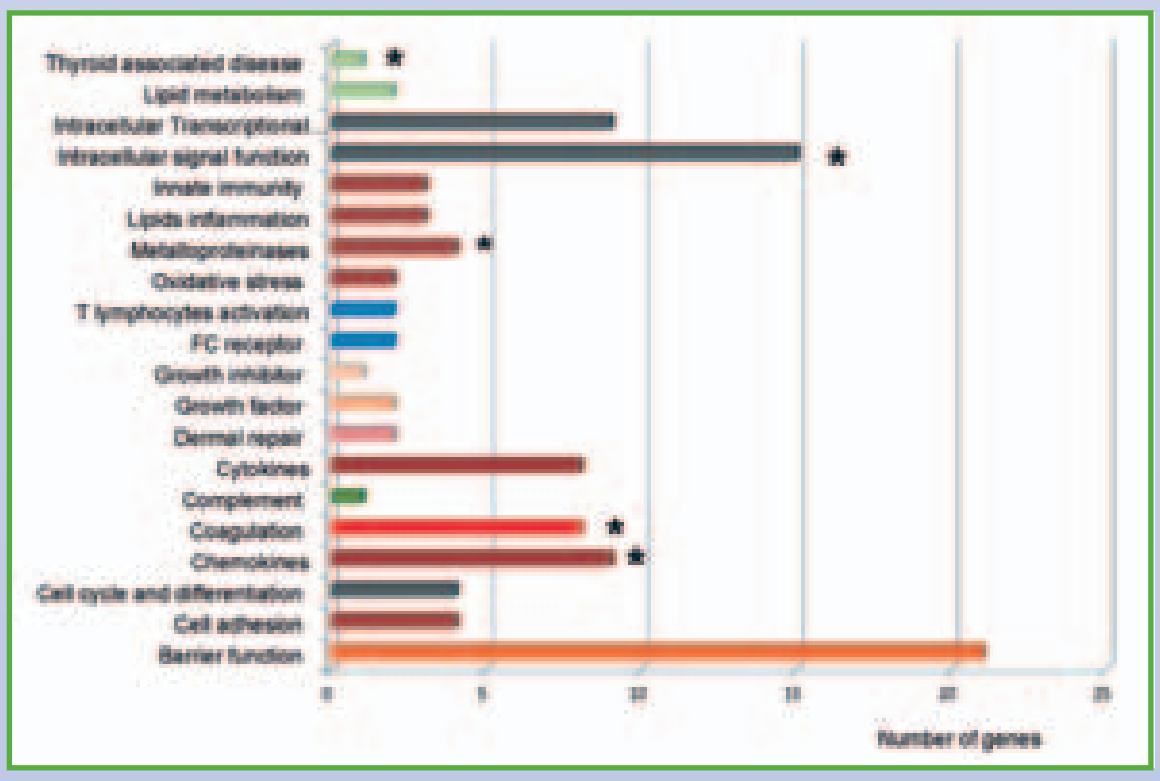
Thyroid associated disease
Lipid metabolism
Intracellular Transcriptional
Intracellular signal function
Innate immunity
Lipids inflammation
Metalloproteinases
Oxidative stress
T lymphocytes activation
FC receptor
Growth inhibitor
Growth factor
Dermal repair
Cytokines
Complement
Coagulation
Chemokines
Cell cycle and differentiation
Cell adhesion
Barrier function
0
5
Number of genes

* Under expressed genes in the wheal confirmed by Q-PCR: LMOD1, CXCL12, PDE5A, VWF, TIMP3, RHPN2, CCL15-CCL14.

CONCLUSIONS AND KEY MESSAGES

The results are robust and consistent according to the strict requirements of the methodology.

Gene printing is completely different in the wheal, compared with the apparently normal skin in patients suffering CSU. Non lesional skin shows a permanent inflammatory status. The most common up regulated genes are involved in the recovery of epidermal barrier, inflammation and dermal repair.

The paired comparison between lesional (wheal) and non lesional skin in patients with CSU shows a broad spectrum of pathogenic genes susceptible to be modulated by the treatment.

A functional analysis according with the clinical characteristics, pathological findings, clinical course of the disease and response to the treatments should help to characterize the phenotypes.

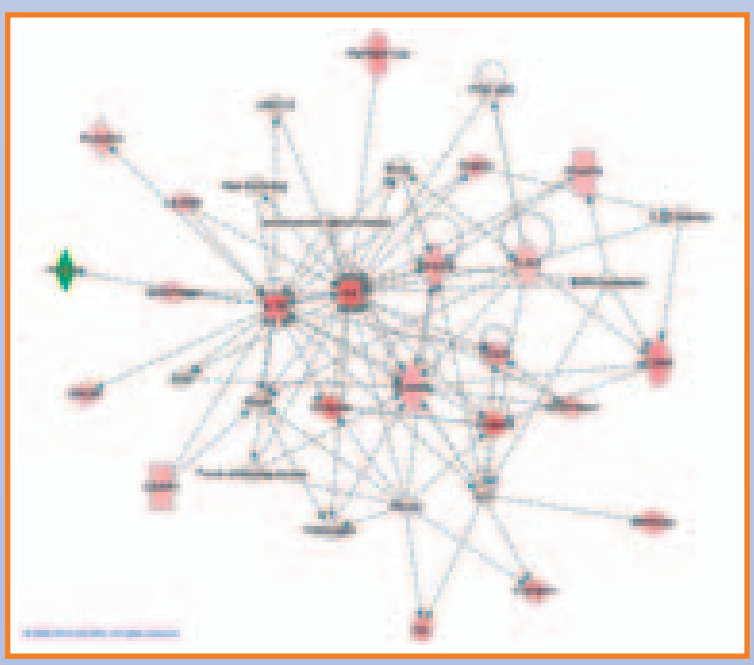

References: Maurer M, Weller K, Bindslev-Jensen C, Giménez-Arnau A, et al. Unmet Clinical needs in chronic spontaneous urticaria. A GA2LEN Task Force Report. Allergy 2011; 66:317-330.

Conflict of Interest: The authors would like to thank Novartis Pharma for the Research Grant that support part of this study.

Hospital del Mar

Parc de Salut MAR Barcelona